Bronchial Allergen Challenge in Asthma: A Model for Inhaled Corticosteroids (ICS) and Montelukast using Literature Summary Data

Farkad Ezzet (1) & Jakob Ribbing (2)

(1) Pharsight, A Certara Company, St Louis, MO, USA, (2) Global Pharmacometrics, Pfizer Limited, Sandwich, Kent, UK

OBJECTIVES

- To characterize effectiveness of anti-asthmatic treatments in studies using bronchial allergen challenge (AC)

- **Tools & Methodology:**
 - Published scientific literature was used to collect information on response to treatment following AC in randomized controlled studies
 - Summary response (mean FEV1 %CFB) from study arms was used to describe treatment effect
 - A model was used to describe the time course of individual treatments

Model

EAR and LAR were modeled using a sum of 2 gamma density functions, if time =< or > δ hours

$$
\begin{aligned}
\%FEV1_{ij}(time) = &- (time \le \delta).S_{1j}.(1-\alpha_i).Gamma(L_1,\ K_1,\ time) \\
&- (time > \delta).S_{2j}.(1-\beta_i).Gamma(L_2,\ K_2,\ time) \\
&+ \varepsilon_{ij}(time)
\end{aligned}
$$

$\%FEV1_{ij}$: observed mean %CFB in $FEV1$ under ith treatment in the jth study

$time$: observation time following allergen inhalation/treatment administration

$Gamma(L,\ K,\ time)$: gamma density function with $L \& K > 0$

δ: time of end of EAR and beginning of LAR

$\alpha_i \in (0,1)$: attenuation during EAR under ith treatment (intercept)

$\beta_i \in (0,1)$: attenuation during LAR under ith treatment (intercept)

$\alpha_i (or\ \beta_i) = \theta_i + \gamma(Dose/median(Dose) - 1)$

S_{kj}: scale parameter for the jth study, $k = 1, 2$

$S_{kj} = S_k.e^{\eta_{kj}}, \eta_{kj}$ is random effect of the jth study, $\eta_{kj} \sim N(0, \sigma^2_{\eta,k})$

$\varepsilon_{ij} \sim N(0, \sigma^2_{\varepsilon})$

Model Summary

	Parameter	Estimate	SE	%CV	p-value
	S1 Placebo	-47.16	2.62	-5.6	< 0.0001
	Mont	0.58	0.03	4.3	< 0.0001
	Bud & D5158	0.11	0.04	34.6	< 0.0001
α	Bec, Cic, Flu & Mom	0.28	0.03	9.4	< 0.0001
	Cic 800	0.21	0.08	36.8	0.01
	Flu dose	0.24	0.06	26.5	< 0.0001
	S2 Placebo	-1267.19	138.74	-10.9	< 0.0001
	Mont	0.65	0.03	5	< 0.0001
	Bud	0.62	0.03	5.6	< 0.0001
	Bec	0.45	0.1	21	< 0.0001
	Flu	0.86	0.05	6.1	< 0.0001
β	Mom	0.58	0.07	12.6	< 0.0001
	D5158	0.08	0.1	125.7	0.43
	Flu dose	0.57	0.17	29.9	< 0.0001
	Mom dose	0.07	0.04	48.2	0.04
	K1	1.33	0.05	3.4	< 0.0001
	K2	4.94	0.4	8	< 0.0001
	L1	1.27	0.09	6.8	< 0.0001
	L2	0.44	0.06	14.5	< 0.0001
	SD eta.S1	0.22	-	-	-
	SD eta.S2	0.35	-	-	-
	SD Error	12.06	-	-	-

studies=22, # observations=682, Log Lik.= -2722.69

Model Predictions

- The model provided a useful tool to compare effectiveness of marketed drugs in these experimental settings. Treatments are found to attenuate EAR and LAR differently, Figure 3.
- Incorporating imprecision in model parameters, prediction intervals of treatment response provide a more realistic characterization of current knowledge of treatment effect and differences, Figure 4.

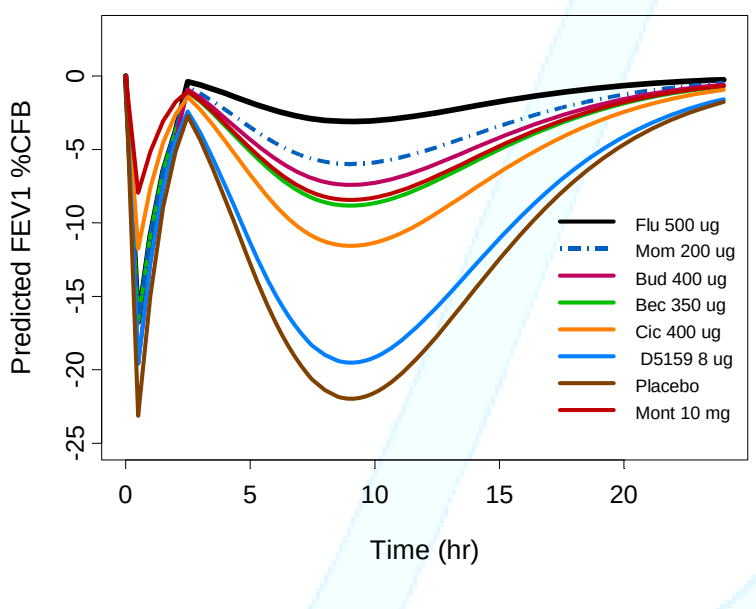
Predicted FEV1 %CFB
Time (hr)
Flu 500 ug
Mom 200 ug
Bud 400 ug
Bec 350 ug
Cic 400 ug
D5159 8 ug
Placebo
Mont 10 mg

Figure 3: Predicted FEV1 %CFB for selected treatment doses

Fluticasone 250 ug and Placebo

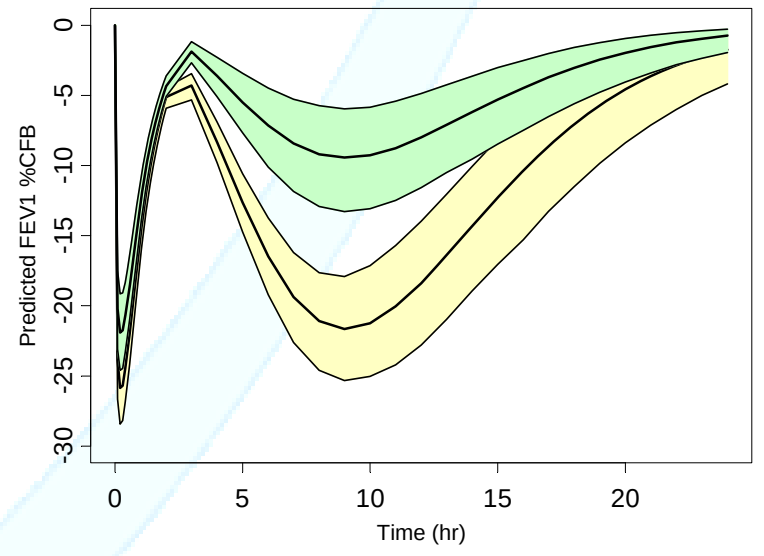
Predicted FEV1 %CFB
Time (hr)

Figure 4: Predicted FEV1 %CFB incorporating model uncertainty for Fluticasone 250 ug and Placebo

BACKGROUND

Data:

Database included 47 randomized double blind placebo controlled studies with FEV information (change from baseline) in mild asthmatic patients. 22 studies used for modeling, 15 on ICS and 7 on Montelukast (Mont). ICS were Budesonide (Bud), Ciclesonide (Cic), Beclomethasone (Bec), Fluticasone (Flu), Mometasone (Mom) and D4158.

Definitions:

- Following AC, patients exhibit early asthmatic response (EAR) and late asthmatic response (LAR), within 30 minutes and 4-8 hours after inhalation of allergen, respectively [1]. Figure 1 shows time course of mean FEV1 %change from baseline (%CFB) under placebo from selected studies
- We sought to estimate %attenuation due to active treatment using placebo as a reference

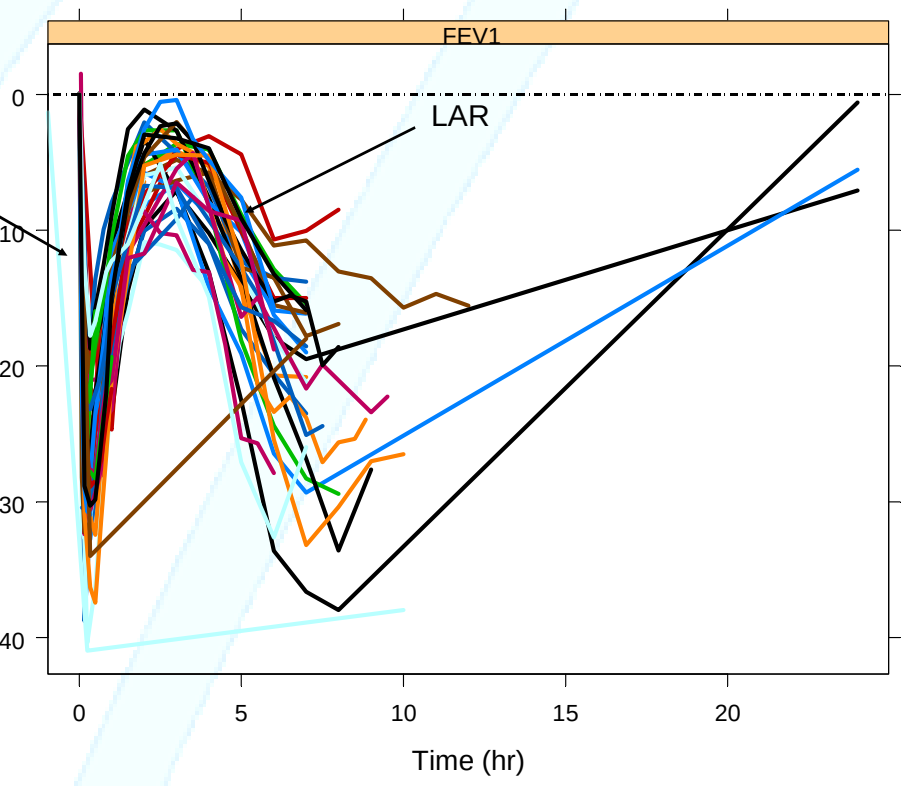
FEV1
EAR
LAR
FEV1 %CFB
Time (hr)

Figure 1: FEV %CFB vs. Time (hr) following AC under placebo EAR & LAR

Model Results Diagnostics and Predictions

- The gamma Model was found suitable in capturing %FEV1. S_1 and S_2, together with attenuation parameters α and β captured most of the differences in %FEV1 between treatments (while L and K were common to all treatments). For example, %attenuation in EAR and LAR for Montelukast (10 mg) was 58% and 65% respectively. For Fluticasone 250 mcg (an ICS) it was 16% and 58%. Budesonide was 11% and 62%.
- The value of δ signifying end of EAR and beginning of LAR was between 2 and 3 hours. A similar value of objective function was achieved for of δ in this range.
- Coefficient of variation of inter-study variability in S_1 and S_2 were 22% and 35%.
- SD of residual error was inflated by square root of sample size, a weighting factor used in model. Accounting for sample size, SD is only 3%.
- Visual predictive checks and posterior predictive checks together with standard diagnostics (Figure 2) indicated adequacy of the model fit. Model estimates were found invariant when subsets of the data were used.

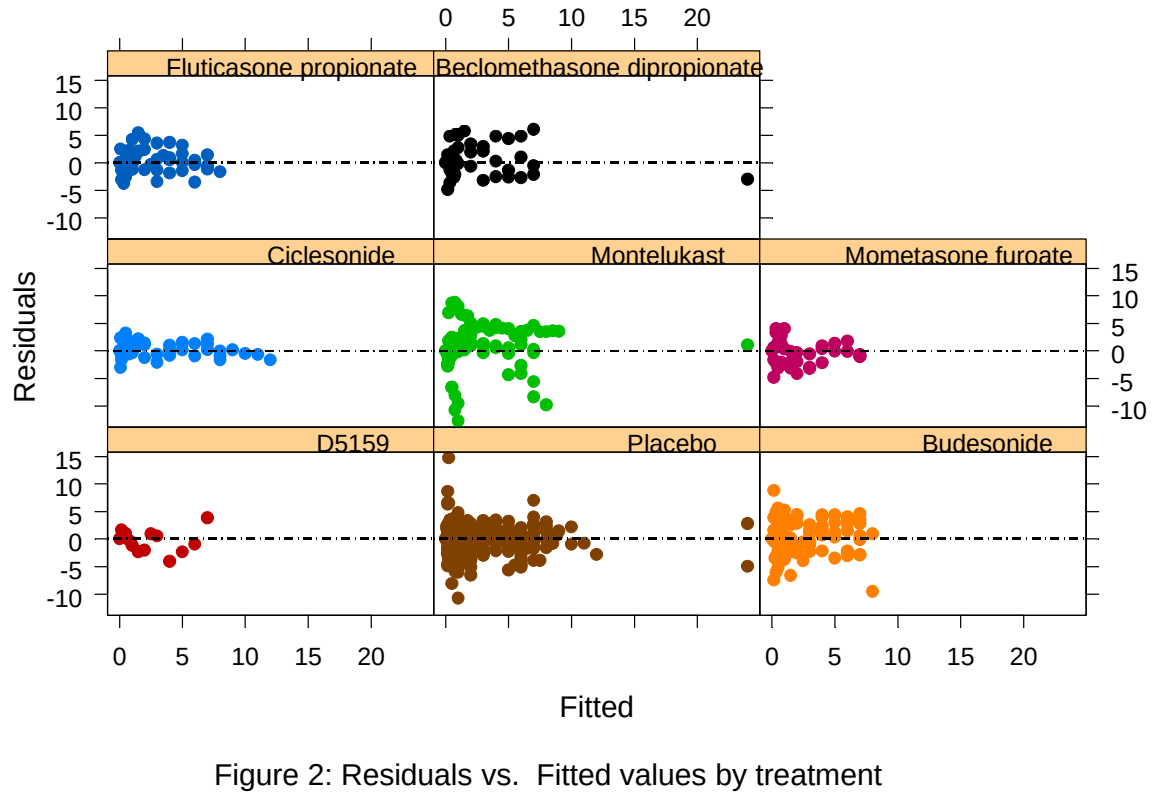
Fluticasone propionate
Beclomethasone dipropionate
Ciclesonide
Montelukast
Mometasone furoate
D5159
Placebo
Budesonide
Residuals
Fitted

Figure 2: Residuals vs. Fitted values by treatment

CONCLUSIONS

- A sum of 2 gamma functions was found to be a flexible model to describing %FEV1 following AC.
- The attenuation parameters α and β captured most of the differences between treatments, allowing a simple and direct comparison.
- The literature model aids the interpretation of ongoing AC studies within Pfizer, as well as design of future AC studies, and can be updated with internal data from positive controls.

REFERENCES

1. Gauvreau GM, Evans MY: Allergen Inhalation Challenge: A Human Model of Asthma Exacerbation. Sjobring U, Taylor JD (eds): Models of Exacerbation in Asthma and COPD. Contrib Microbiol. Basel, Karger, 2007, vol 14, pp 21-32.